COM701 (A NOVEL IMMUNE CHECKPOINT INHIBITOR) IN PATIENTS WITH ADVANCED SOLID TUMORS.

A. ElNaggar[1], J. Luke*[2], E. Hamilton[3], B. Chmielowski[4], E. Ileana-Dumbrava[5], A. Patnaik[6], E. Lim[7], A.H Adewoye[8], J. Hunter[8], J. Olweny[8], M. Sharma[9], R. Sullivan[10], D. Vaena[1], D. Rasco[6].

[1]West Cancer Center and Research Institute., Gynecologic Oncology., Memphis, TN., USA. [2]The University of Chicago Medical Center., Medical Oncology., Chicago, IL., USA. [3]Sarah Cannon Research Institute/Tennessee Oncology., Medical Oncology., Nashville, TN., USA. [4]University of California Los Angeles., Medical Oncology, Los Angeles, CA., USA. [5]The University of Texas MD Anderson Cancer Center., Department of Investigational Cancer Therapeutics., Houston, TX., USA. [6]South Texas Accelerated Research Therapeutics START., Medical Oncology., San Antonio, TX., USA. [7]Columbia University., Medical Oncology., New York, NY., USA. [8]Compugen USA Inc., Clinical Development., South San Francisco, CA, USA. [9]START Midwest., Medical Oncology., Grand Rapids, MI., USA. [10]Massachusetts General Hospital., Medical Oncology., Boston, MA., USA.

*Now at University of Pittsburgh Medical Center; Medical Oncology., Pittsburgh, PA; USA.

Objectives:

COM701 is a novel 1st in-class monoclonal antibody (checkpoint inhibitor) of poliovirus receptor related immunoglobulin domain containing (PVRIG) blocking its interaction with it's ligand, PVRL2 (Fig 1). In preclinical experiments inhibition of PVRIG leads to activation of T-cells in the tumor microenvironment generating an anti-tumor immune response leading to tumor growth inhibition (Fig 2). We hypothesized that COM701 monotherapy and in combination with nivolumab will be safe and tolerable and demonstrate antitumor activity in pts with advanced solid tumors. Key primary objectives: safety and tolerability of COM701 monotherapy and in combination with nivolumab measured by the incidence of pts with adverse events and dose-limiting toxicities. The recommended dose for expansion of COM701 monotherapy and in combination with nivolumab. Key secondary objectives: the preliminary antitumor activity of COM701 monotherapy and with nivolumab in pts with endometrial, ovarian, breast and lung cancer. These tumor types were selected based on high expression of PVRL2.

Methods:

Study design: Dose escalation hybrid accelerated titration and 3+3 design. Key inclusion criteria: Age ≥18 yrs, advanced solid tumor and has exhausted all available standard therapy, prior checkpoint inhibitor permissible. Key exclusion criteria: history of immune-related events that led to immunotherapy treatment discontinuation. Expansion cohorts will enroll pts with the tumor types selected. ClinicalTrials.gov Identifier: NCT03667716.

Results:

No DLTs have been observed up to 7th COM701 monotherapy dose level and dose level 2 in Arm B (Fig 3).

Conclusions:

COM701 monotherapy and in combination with nivolumab has an acceptable safety and tolerable profile at the dose levels tested. Study enrollment is ongoing.

Fig 1.

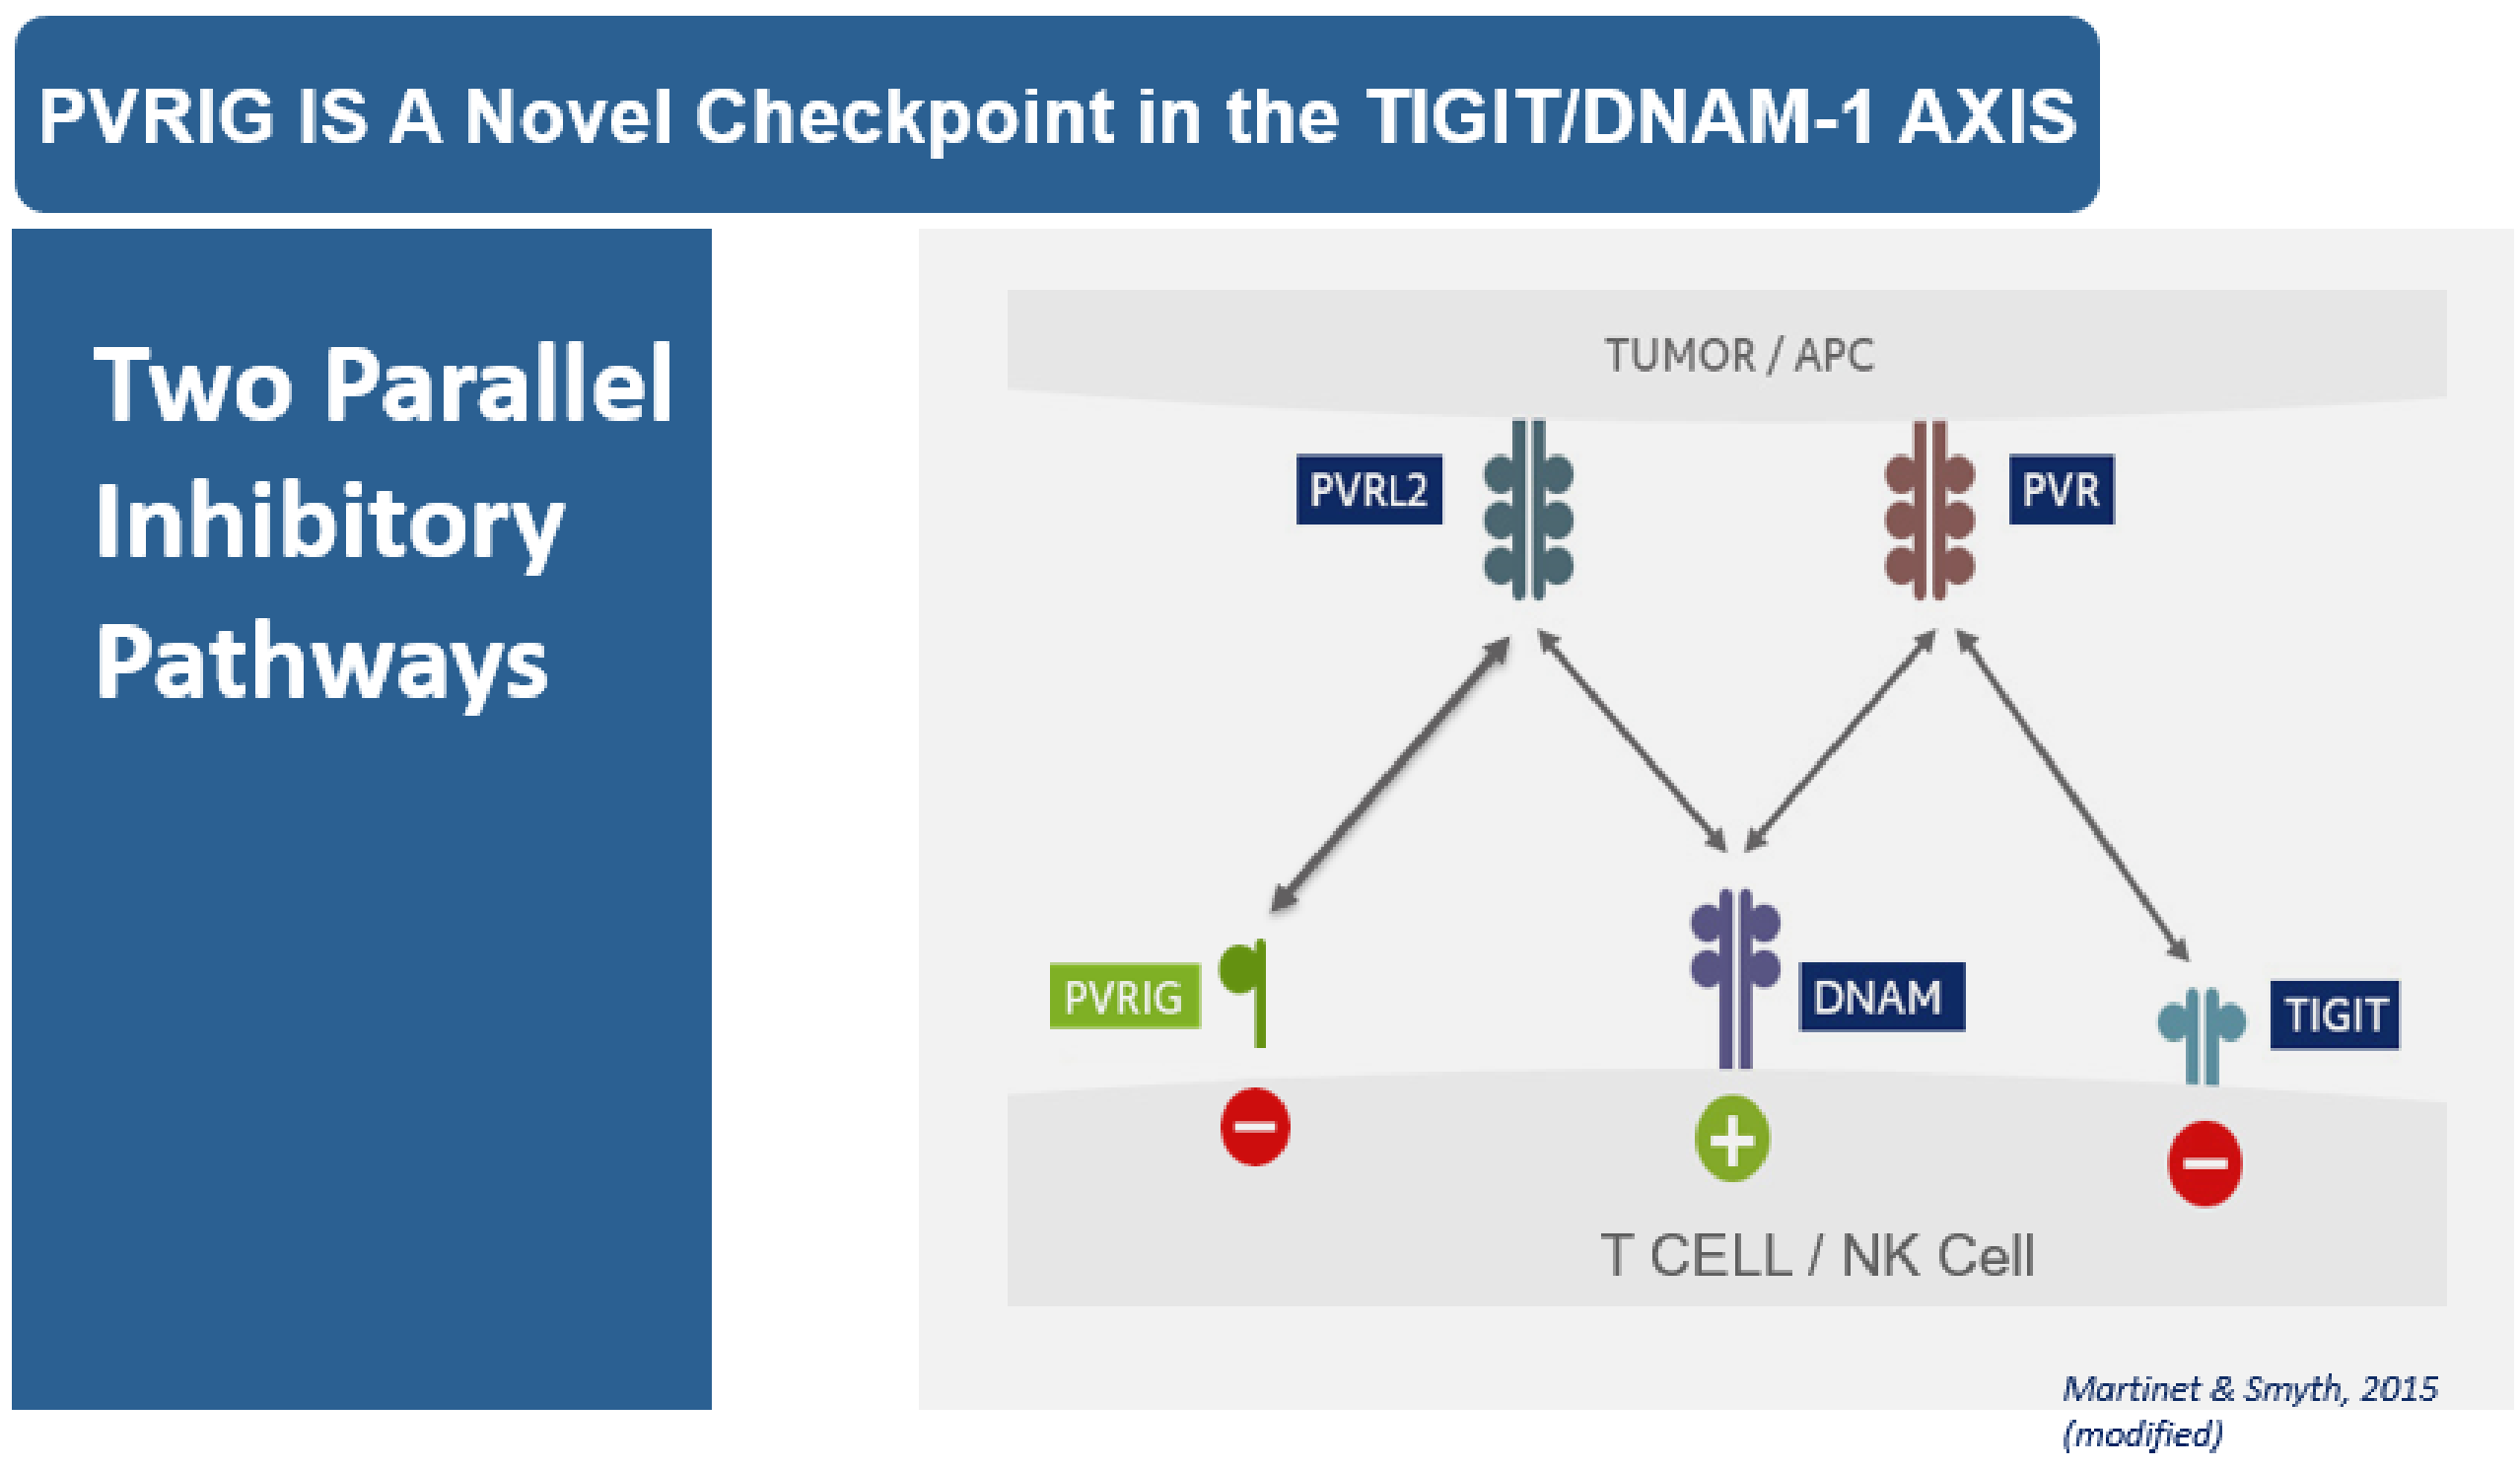
PVRIG IS A Novel Checkpoint in the TIGIT/DNAM-1 AXIS
Two Parallel Inhibitory Pathways
TUMOR / APC
PVRL2
PVR
PVRIG
DNAM
TIGIT
T CELL / NK Cell
Martinet & Smyth, 2015 (modified)

Fig 2.

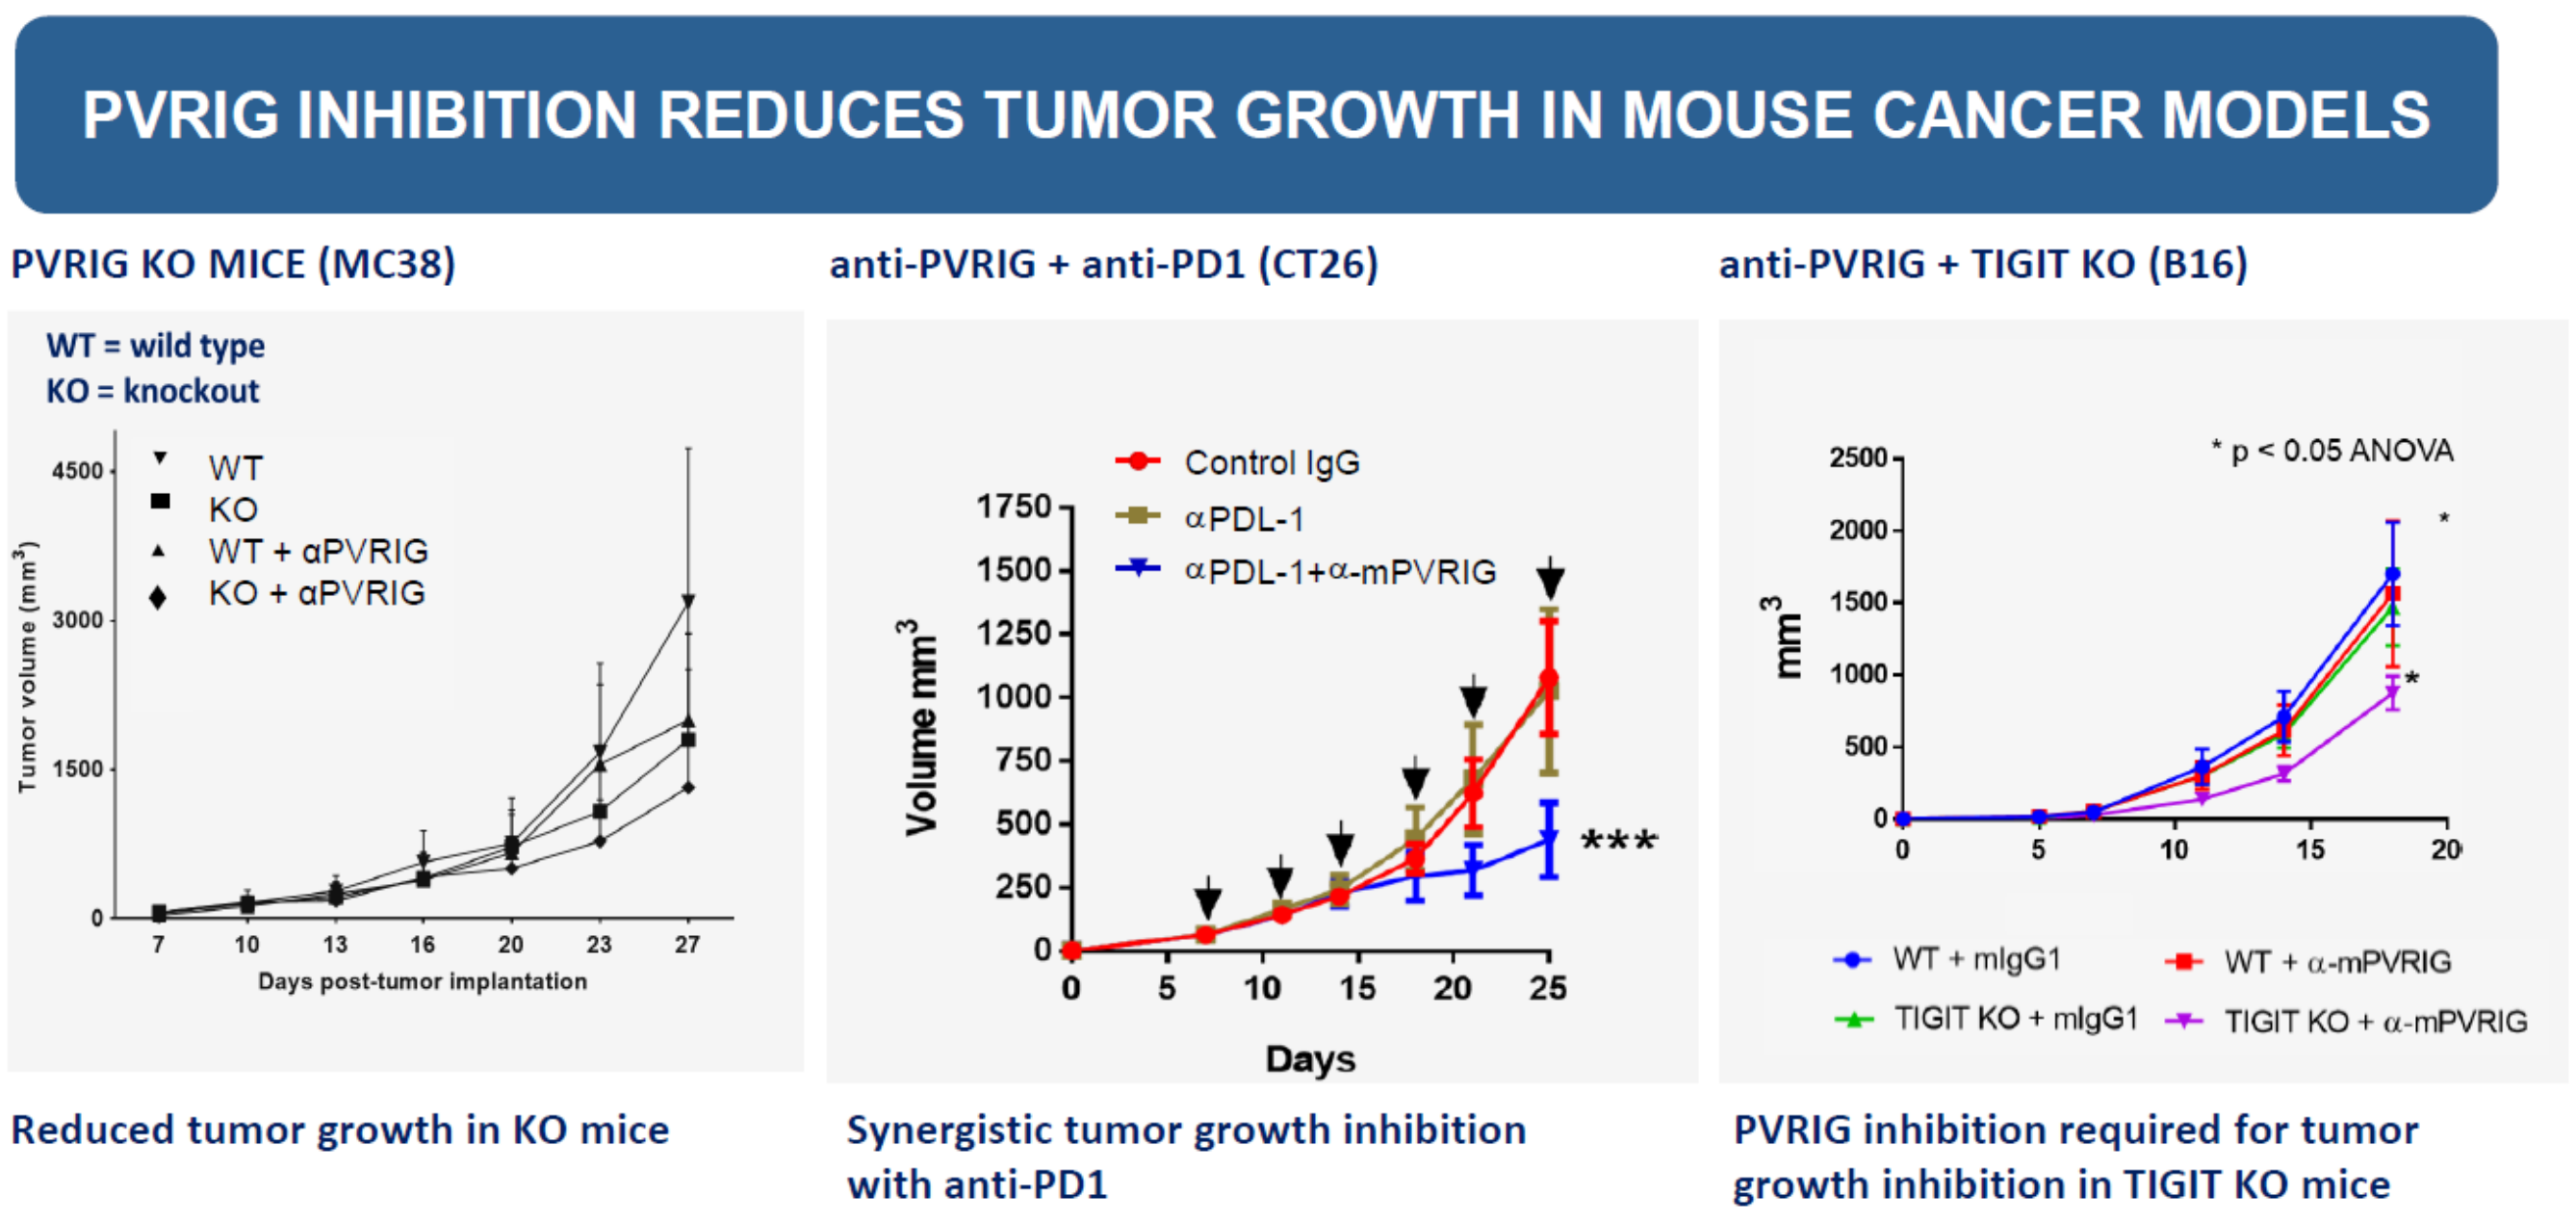
PVRIG INHIBITION REDUCES TUMOR GROWTH IN MOUSE CANCER MODELS
PVRIG KO MICE (MC38)
WT = wild type
KO = knockout
WT
KO
WT + αPVRIG
KO + αPVRIG
Tumor volume (mm³)
Days post-tumor implantation
Reduced tumor growth in KO mice
anti-PVRIG + anti-PD1 (CT26)
Control IgG
αPDL-1
αPDL-1+α-mPVRIG
Volume mm³
Days

Synergistic tumor growth inhibition with anti-PD1
anti-PVRIG + TIGIT KO (B16)
* p < 0.05 ANOVA
mm³
WT + mIgG1
WT + α-mPVRIG
TIGIT KO + mIgG1
TIGIT KO + α-mPVRIG
PVRIG inhibition required for tumor growth inhibition in TIGIT KO mice

Fig 3.

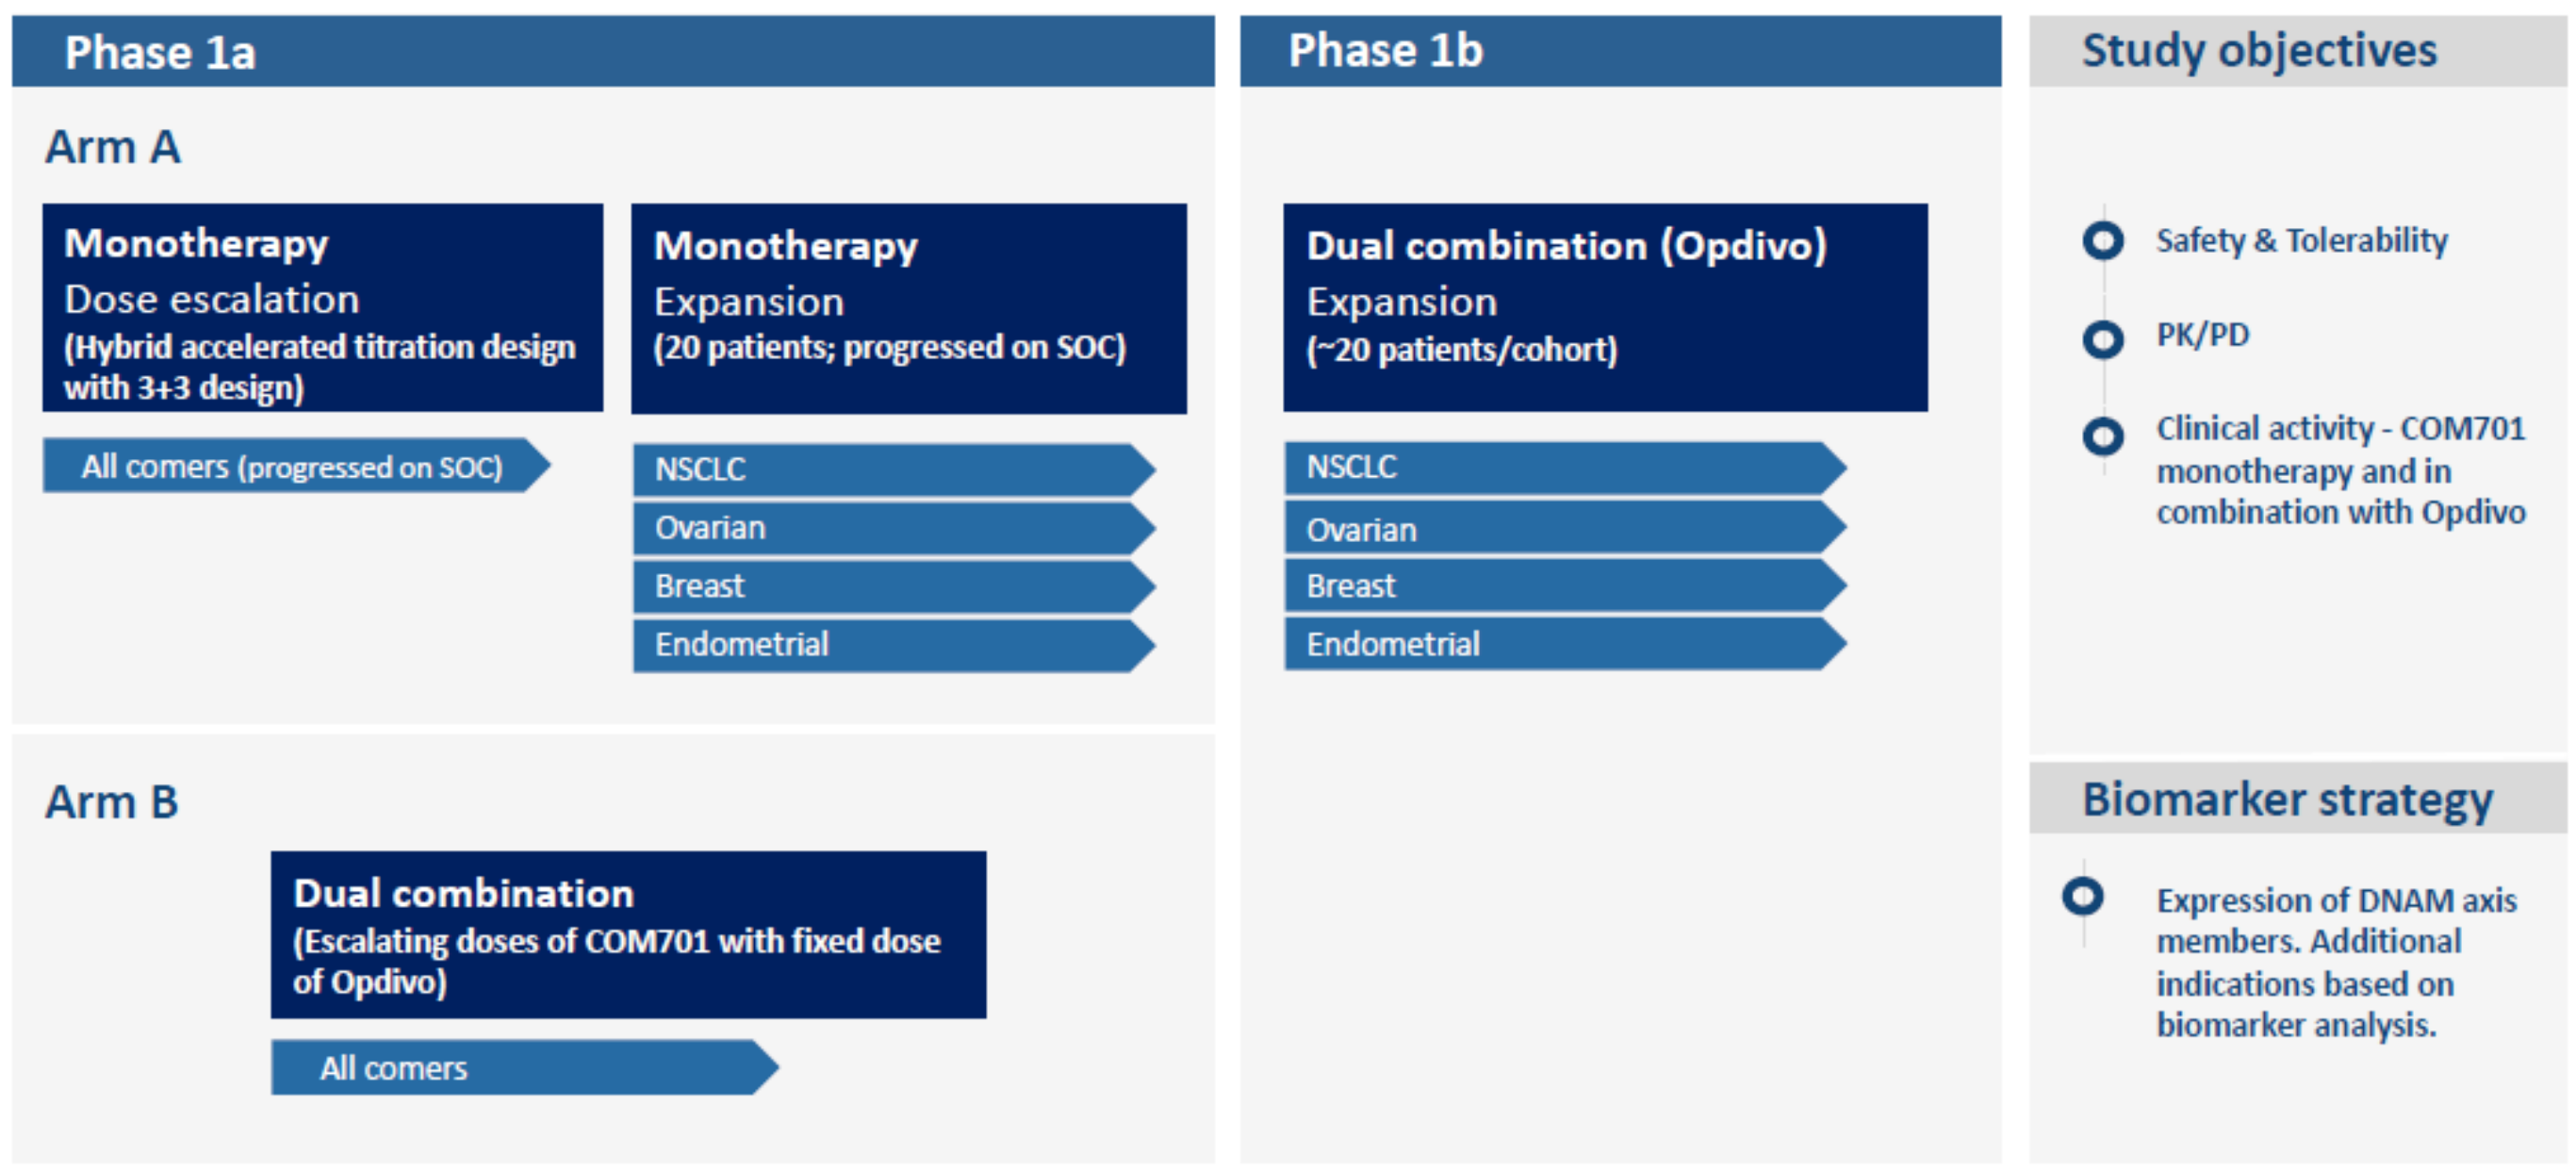
Phase 1a
Arm A
Monotherapy
Dose escalation
(Hybrid accelerated titration design with 3+3 design)
All comers (progressed on SOC)
Monotherapy
Expansion
(20 patients; progressed on SOC)
NSCLC
Ovarian
Breast
Endometrial
Arm B
Dual combination
(Escalating doses of COM701 with fixed dose of Opdivo)
All comers
Phase 1b
Dual combination (Opdivo)
Expansion
(~20 patients/cohort)
NSCLC
Ovarian
Breast
Endometrial
Study objectives
Safety & Tolerability
PK/PD
Clinical activity - COM701 monotherapy and in combination with Opdivo
Biomarker strategy
Expression of DNAM axis members. Additional indications based on biomarker analysis.

Acknowledgment: We thank the patients and their families, clinical trial investigators and their staff for participating in this clinical trial.

Study NCT03667716 is in collaboration with Bristol-Myers Squibb.

Study sponsored by Compugen Ltd.
For more information: www.cgen.com

Copyright © 2019 Presented at International Gynecologic Cancer Society Annual Global Meeting Rio de Janeiro, Brazil Sept 19 – 21 2019.